Poster 40

Gene and Protein Expression Of MAGE, PD-L1 and Associated Immune Landscape Elements in Non-small Cell Lung Carcinoma, Urothelial Carcinoma, Squamous Cell Cancer Of The Head And Neck, and Cervical Carcinoma

Stephanie H. Astrow[1], Izak Faiena[2], Rajul K. Jain[1], Alexandra Drakaki[2], Wesley S.Chang[3], Clark C. Fjeld[3], Jin Li[4], and Adrian Bot[1]

[1]Kite, a Gilead company, Santa Monica, CA; [2]University of California, Los Angeles, CA; [3]Molecular MD, Portland, OR; [4]Molecular MD, Cambridge,MA

BACKGROUND

- Therapeutic options are limited for patients who have advanced solid tumors that progress on conventional chemotherapy
- Clinical responses have been observed with adoptively transferred tumor-specific T cells, including a T-cell receptor (TCR)-based therapy targeting the cancer testis antigens melanoma-associated antigen 3 (MAGE-A3) and melanoma-associated antigen 6 (MAGE-A6)[1]
 - A T-cell product with this TCR is currently being tested in a novel phase 1 multicenter trial (NCT03139370)
 - This TCR is major histocompatibility complex (MHC) class II restricted and recognizes peptide in the context of HLA DPB1*04:01
- The expression of MAGE-A antigens is prevalent in diverse solid tumors[2]
- The intratumor expression of MAGE-A3/A6 and its localization in relation to programmed death ligand 1 (PD-L1) are of interest as they may create a rationale for MAGE-A3/A6–directed TCR therapy, alone or in conjunction with PD-L1 inhibition
- We developed a 2-part assay system combining immunohistochemistry (IHC) and reverse-transcription quantitative polymerase chain reaction (RT-qPCR) to detect MAGE-A3/A6 expression
- A number of non-small cell lung cancer (NSCLC), urothelial carcinoma (UC), squamous cell carcinoma of the head and neck (SCCHN), and cervical cancer (CC) tumors were profiled for MAGE-A3/A6
 - In addition, PD-L1 and MHC II were profiled by IHC, and other immune-related genes were profiled using RNA

METHODS

IHC

- Sections from formalin-fixed, paraffin-embedded tumor specimens were stained by IHC with monoclonal antibodies for:
 - MAGE-A (clone 6C1; recognizes MAGE-A1, A2, A3, A4, A6, A10, A12)
 - PD-L1 (clone E1L3N)
 - MHC II (clone CR43/3)
- Staining was performed on the Bond III platform (Leica Biosystems), using Polymer Refine Detection kit
- Slides were scanned into a Aperio AT2 whole-slide scanning system (Leica Biosystems)
 - Regions of interest were captured using Aperio ImageScope software (Leica Biosystems)

RT-qPCR

- MAGE-A3/A6 expression analysis was performed with a validated 2-step RT-qPCR assay specific for MAGE-A3 and-A6
 - The validation showed no cross-reactivity to closely related MAGE-A family members, and amplified products were verified by Sanger sequencing
- Two reference gene assays were used to evaluate RNA quality and to qualify samples
- A quantification cycle (Cq) cutoff was used to make calls (rather than a ΔCq cutoff)
- Positive, negative, and no template controls were analyzed with each run to ensure assay performance

RNA SEQUENCING

- Ninety-six samples were analyzed using the Oncomine™ Immune Response Research Assay (OIRRA; Thermo Fisher Scientific)
 - This targeted RNA sequencing panel measures the expression of 391 genes
 - RNA quality was assessed using a custom RT-qPCR assay measuring the expression of 5 housekeeping genes in a single reaction
 - RNA (10 ng) passing quality control was processed for OIRRA library prep by following the standard protocol
 - Mixture of barcoded libraries from 32 samples was clonally amplified using Ion Chef, loaded onto a 540 chip, and sequenced on the Ion S5™ XL system (Thermo Fisher Scientific)
 - Data analysis was performed using Torrent Suite 5.4 (Thermo Fisher Scientific), excel, DNA-Chip Analyzer, and pheatmap in R package

Figure 1. Study design.

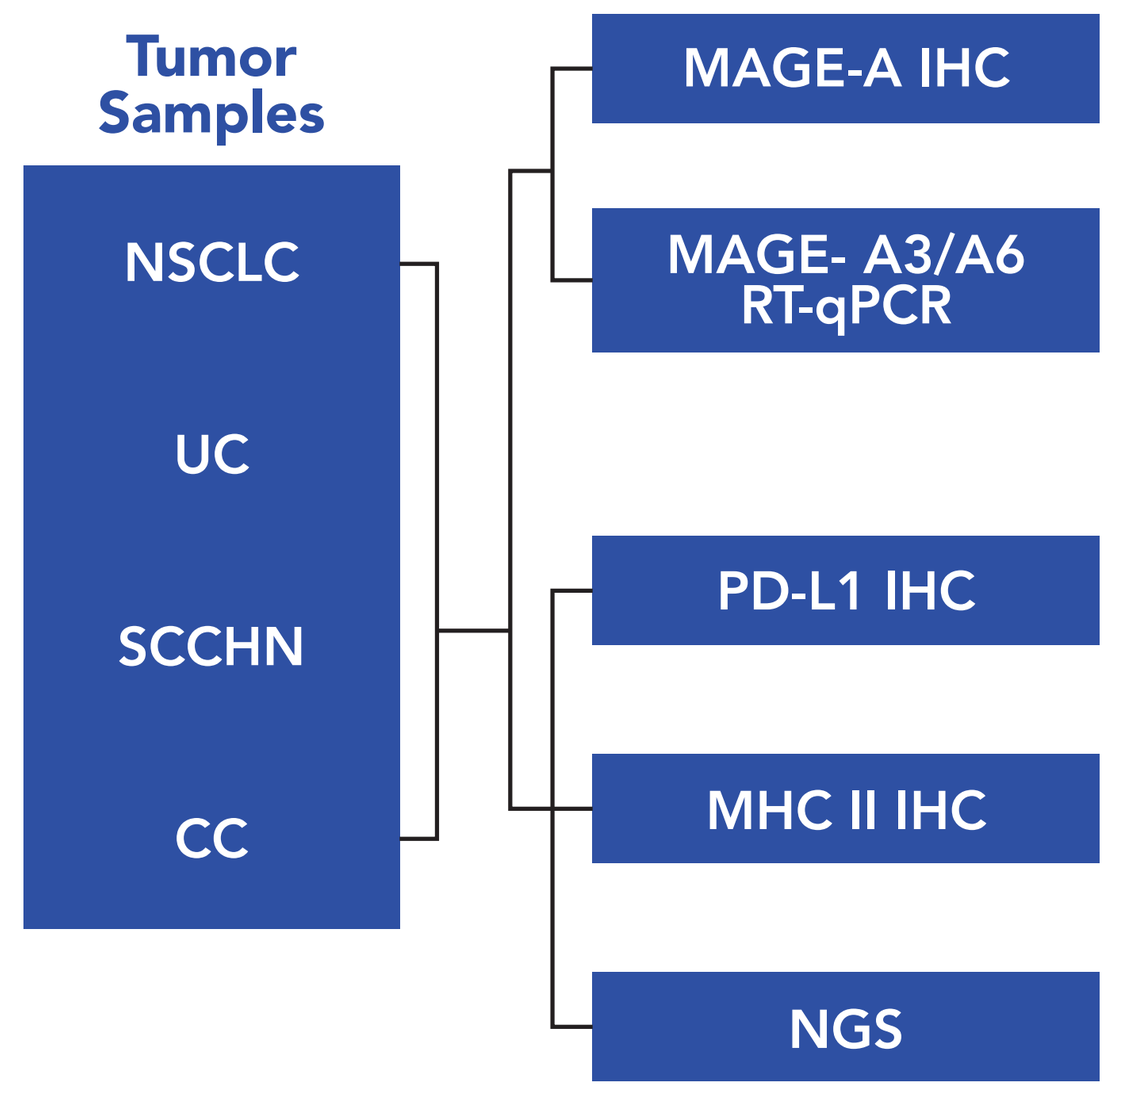

Tumor Samples
NSCLC
UC
SCCHN
CC
MAGE-A IHC
MAGE- A3/A6 RT-qPCR
PD-L1 IHC
MHC II IHC
NGS

A set of 194 NSCLC, UC, SCCHN, and CC tumors were tested with IHC for MAGE-A and RT-qPCR for MAGE-A3/A6. Adjacent sections of a subset of the samples were examined for PD-L1 and MHC II by IHC and for immune-related gene expression by next-generation sequencing.
CC, cervical cancer; IHC, immunohistochemistry; MAGE, melanoma-associated antigen; MHC, major histocompatibility complex; NGS, next-generation sequencing; NSCLC, non-small cell lung cancer; PD-L1, programmed death ligand 1; RT-qPCR, reverse-transcription quantitative polymerase chain reaction; SCCHN, squamous cell carcinoma of the head and neck; UC, urothelial carcinoma.

RESULTS

Table 1. A combination of MAGE-A–positive IHC and MAGE-A3/A6 RT-qPCR can be used to select patients with MAGE-A3/A6–positive tumors.

TumorType	N	MAGE-A–positive IHC, n (%)	MAGE-A3/A6 RT-qPCR, n (%)	MAGE-A–positive IHC ≥ cutoff, n (%)	Positive screening test[a], n (%)
NSCLC	81	41 (50)	36 (44)	25 (31)	20 (25)
UC	67	43 (64)	37 (55)	29 (43)	22 (33)
SCCHN	25	17 (68)	14 (56)	12 (48)	11 (44)
CC	21	7 (33)	3 (14)	2 (10)	1 (5)
Total	194	108 (56)	90 (46)	68 (35)	54 (28)

[a]Positive by both RT-qPCR AND IHC ≥ cutoff.
CC, cervical cancer; IHC, immunohistocompatibility; MAGE, melanoma-associated antigen; NSCLC, non-small cell lung cancer; OIRRA, Oncomine™ Immune Response Research Assay; RT-qPCR, reverse-transcription quantitative polymerase chain reaction; SCCHN, small cell carcinoma of the head and neck; TCR, T-cell receptor; UC, urothelial carcinoma.

- Analytical validation of the MAGE-A3/A6 assay system successfully met predefined acceptance criteria appropriate for clinical sample testing
- Consistent with previous trials using MAGE-A IHC to select candidates for therapy with this TCR, the screening assay cutoff is defined as ≥ 50% tumor cells staining with 2+ or 3+ intensity
- For MAGE A3/A6 RT-qPCR, the screening cutoff for a positive result is a Cq < 37
- OIRRA analysis of MAGE-A3/6 expression further confirmed the qPCR expression result, with 94% concordance and coefficient of determination at 0.87

Figure 2. Most MAGE-A–positive tumors express MAGE-A3/A6.

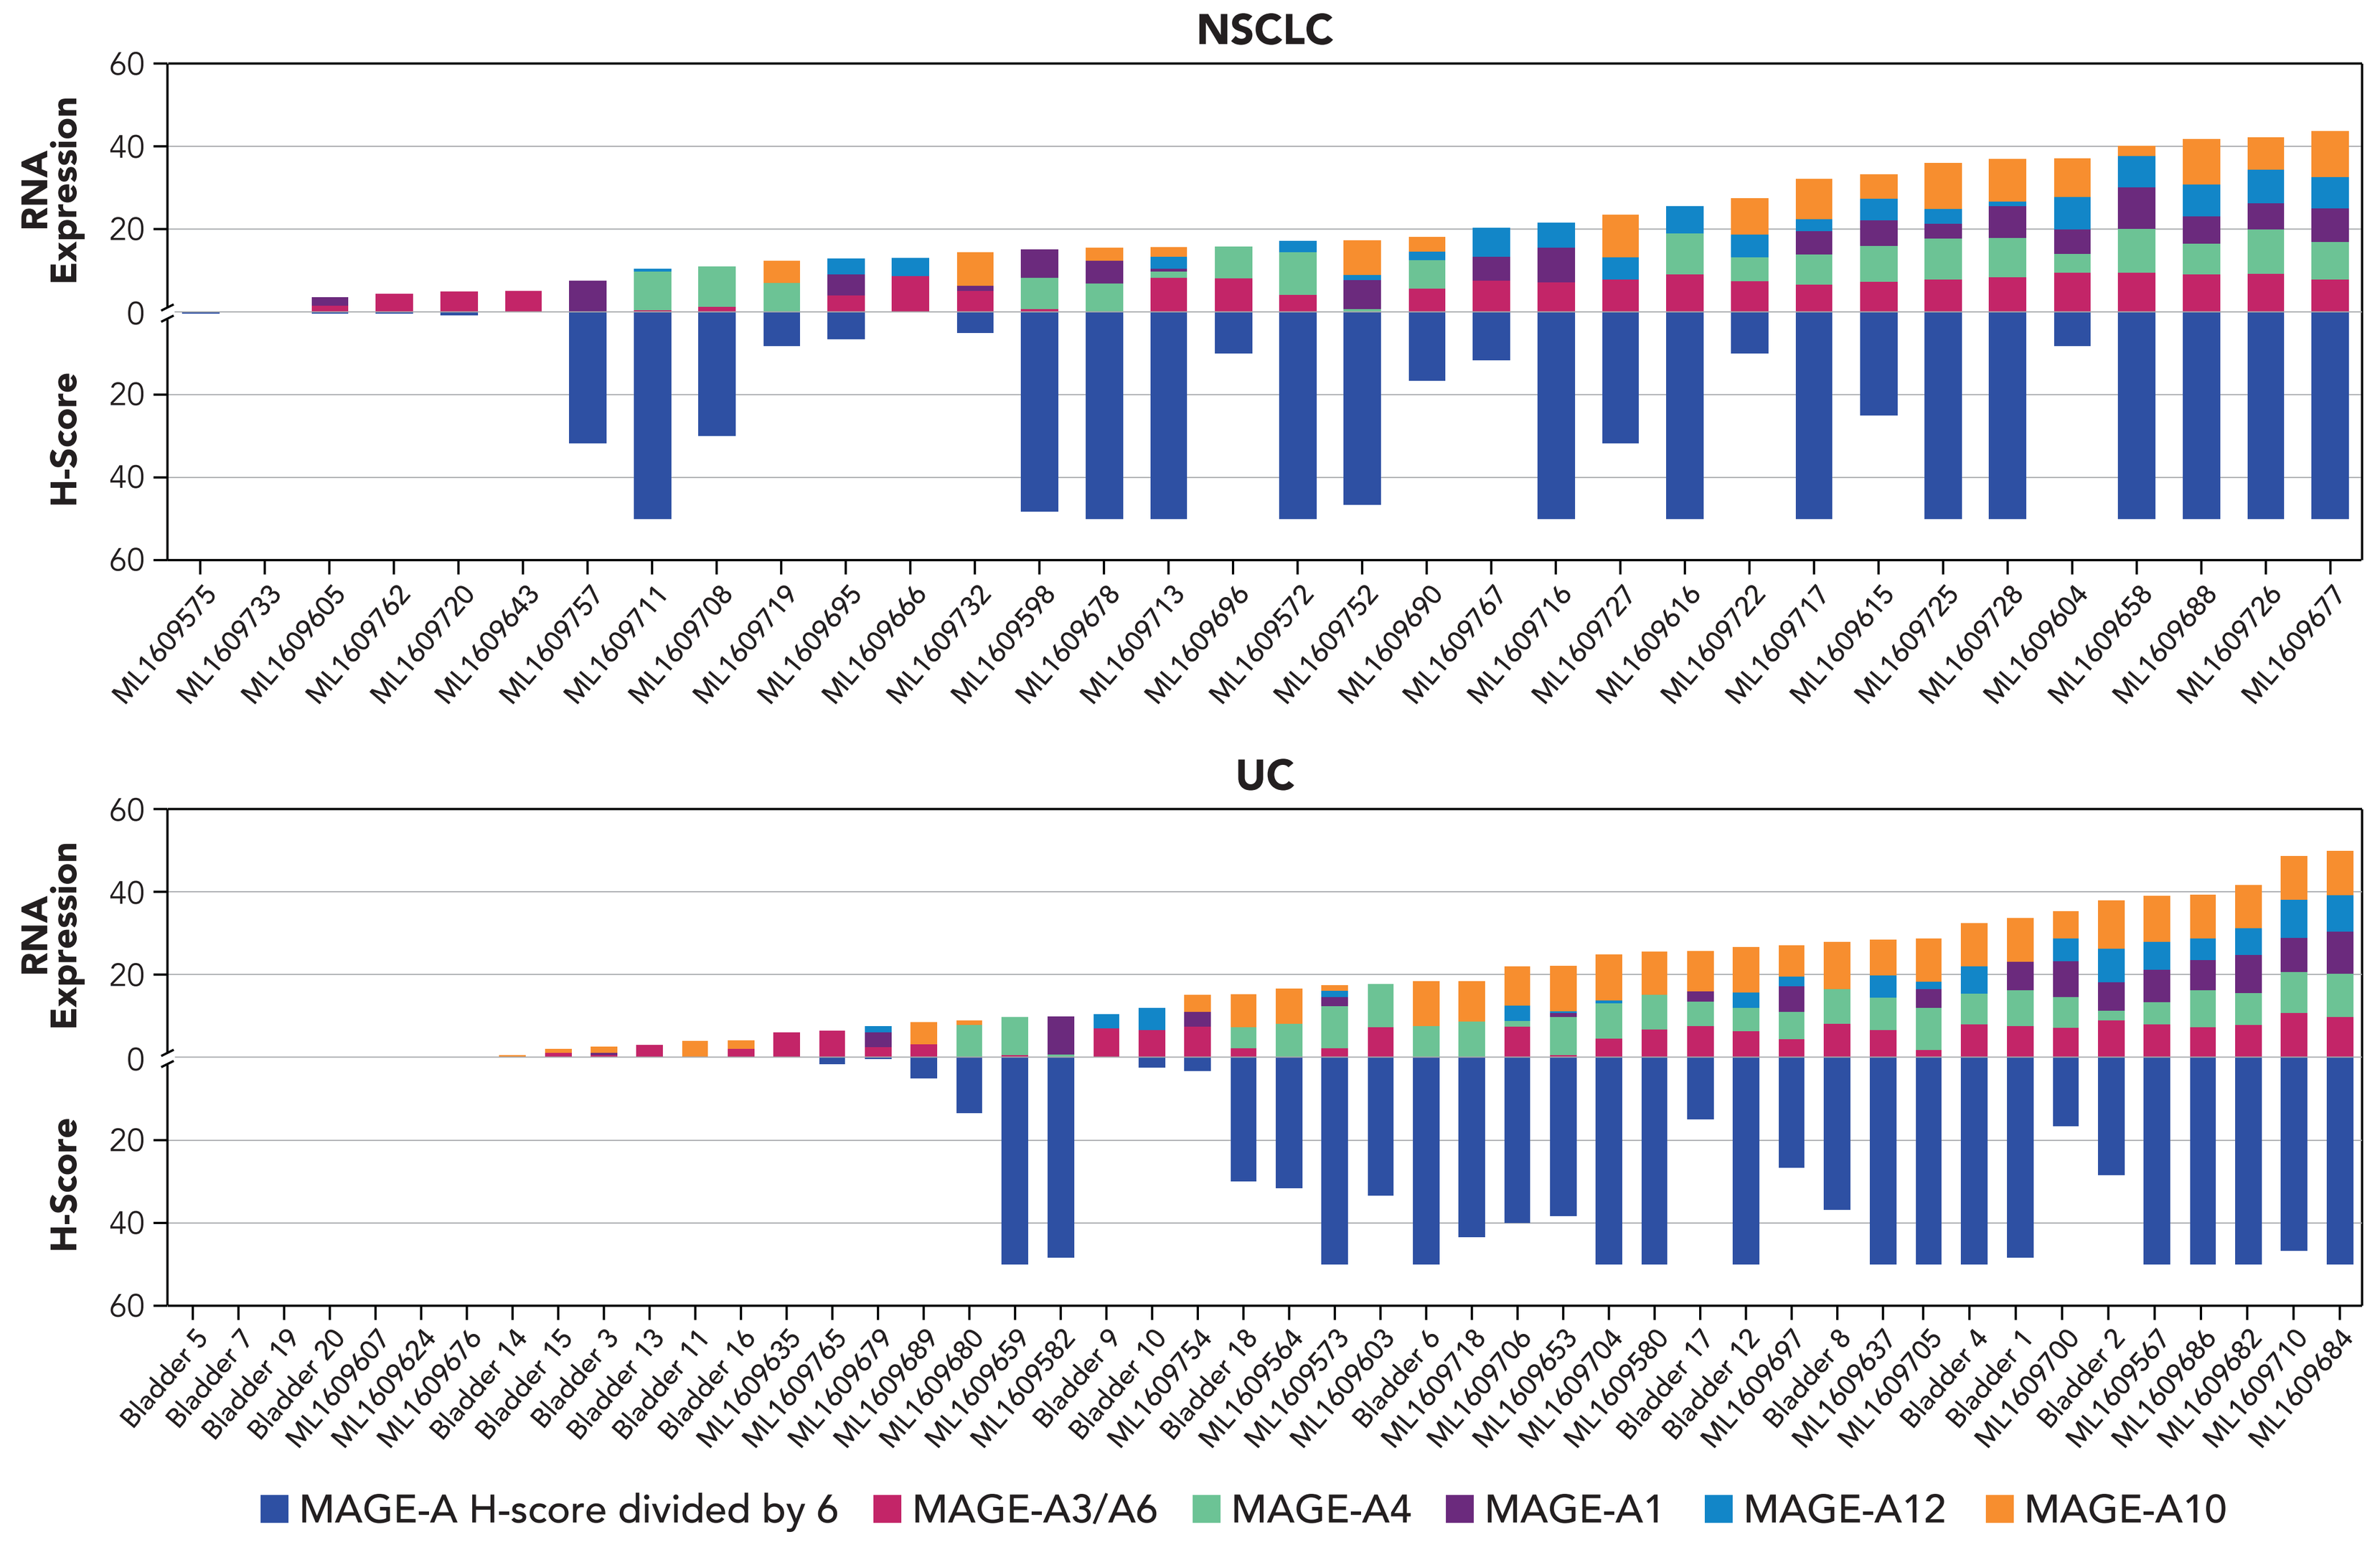

NSCLC
RNA Expression
H-Score
UC
RNA Expression
H-Score
MAGE-A H-score divided by 6
MAGE-A3/A6
MAGE-A4
MAGE-A1
MAGE-A12
MAGE-A10

Using OIRRA, expression of MAGE-A3/A6, as well as the other MAGE family members that are potential contributors to the mAb 6C1 signal (MAGE-A1, A4, A10, and A12) were normalized to RPM and transformed to LOG2 (RPM+1) format. UC and NSCLC samples were sorted using the total expression of MAGE-A1,3/6,4,10,12 by OIRRA (MAGE-A IHC H-score expressed here as a negative value divided by 6 for easy viewing). MAGE transcripts are detected by next-generation sequencing in some samples that are negative by IHC; this is likely because of differences in the sensitivity of the technologies.
mAb, monoclonal antibody; MAGE, melanoma-associated antigen; NSCLC, non-small cell lung cancer; OIRRA, Oncomine™ Immune Response Research Assay; PD-L1, programmed death ligand 1; RPM, reads per million of mapped reads; Th1, type 1 T helper; Treg, regulatory T; UC, urothelial carcinoma.

Table 2. PD-L1 and MHC II can be expressed in both tumor cells and immune cells in MAGE-A3/A6–positive tumors.

	NSCLC (N = 20)	UC (N = 22)	SCCHN (N = 11)
PD-L1 Expression, n (%)			
Tumor or immune cells	18 (90)	10 (45)	10 (91)
Tumor cells	9 (45)	5 (23)	8 (73)
Immune cells	17 (85)	8 (36)	10 (91)
MHC II, n (%)			
Tumor cells	5 (25)	8 (36)	5 (45)

MAGE, melanoma-associated antigen; IHC, immunohistochemistry; MHC, major histocompatibility complex; NSCLC, non-small cell lung cancer; PD-L1, programmed death ligand 1; SCCHN, squamous cell carcinoma of the head and neck; UC, urothelial carcinoma.

- Constitutive expression of MHC class II molecules is restricted to antigen-presenting cells such as dendritic cells, B-lymphocytes, and macrophages
 - MHC II expression can be induced by interferon-γ in most nonimmune cells[3]
- PD-L1 and MHC II expression was examined for MAGE-A3/A6–positive tumors by IHC, with tumor cells and immune cells analyzed separately
 - Approximately 90% of the tumor samples of NSCLC and SCCHN had PD-L1–positive tumor or immune cells, whereas PD-L1 expression was 45% for UC
 - All tumors contained MHC-II–positive immune cells (not shown), whereas the expression of MHC II in tumor cells varied from 25%-45% across histologies

Figure 3. MAGE-A, PD-L1, and MHC II IHC show differential staining patterns with areas of both overlap and exclusivity.

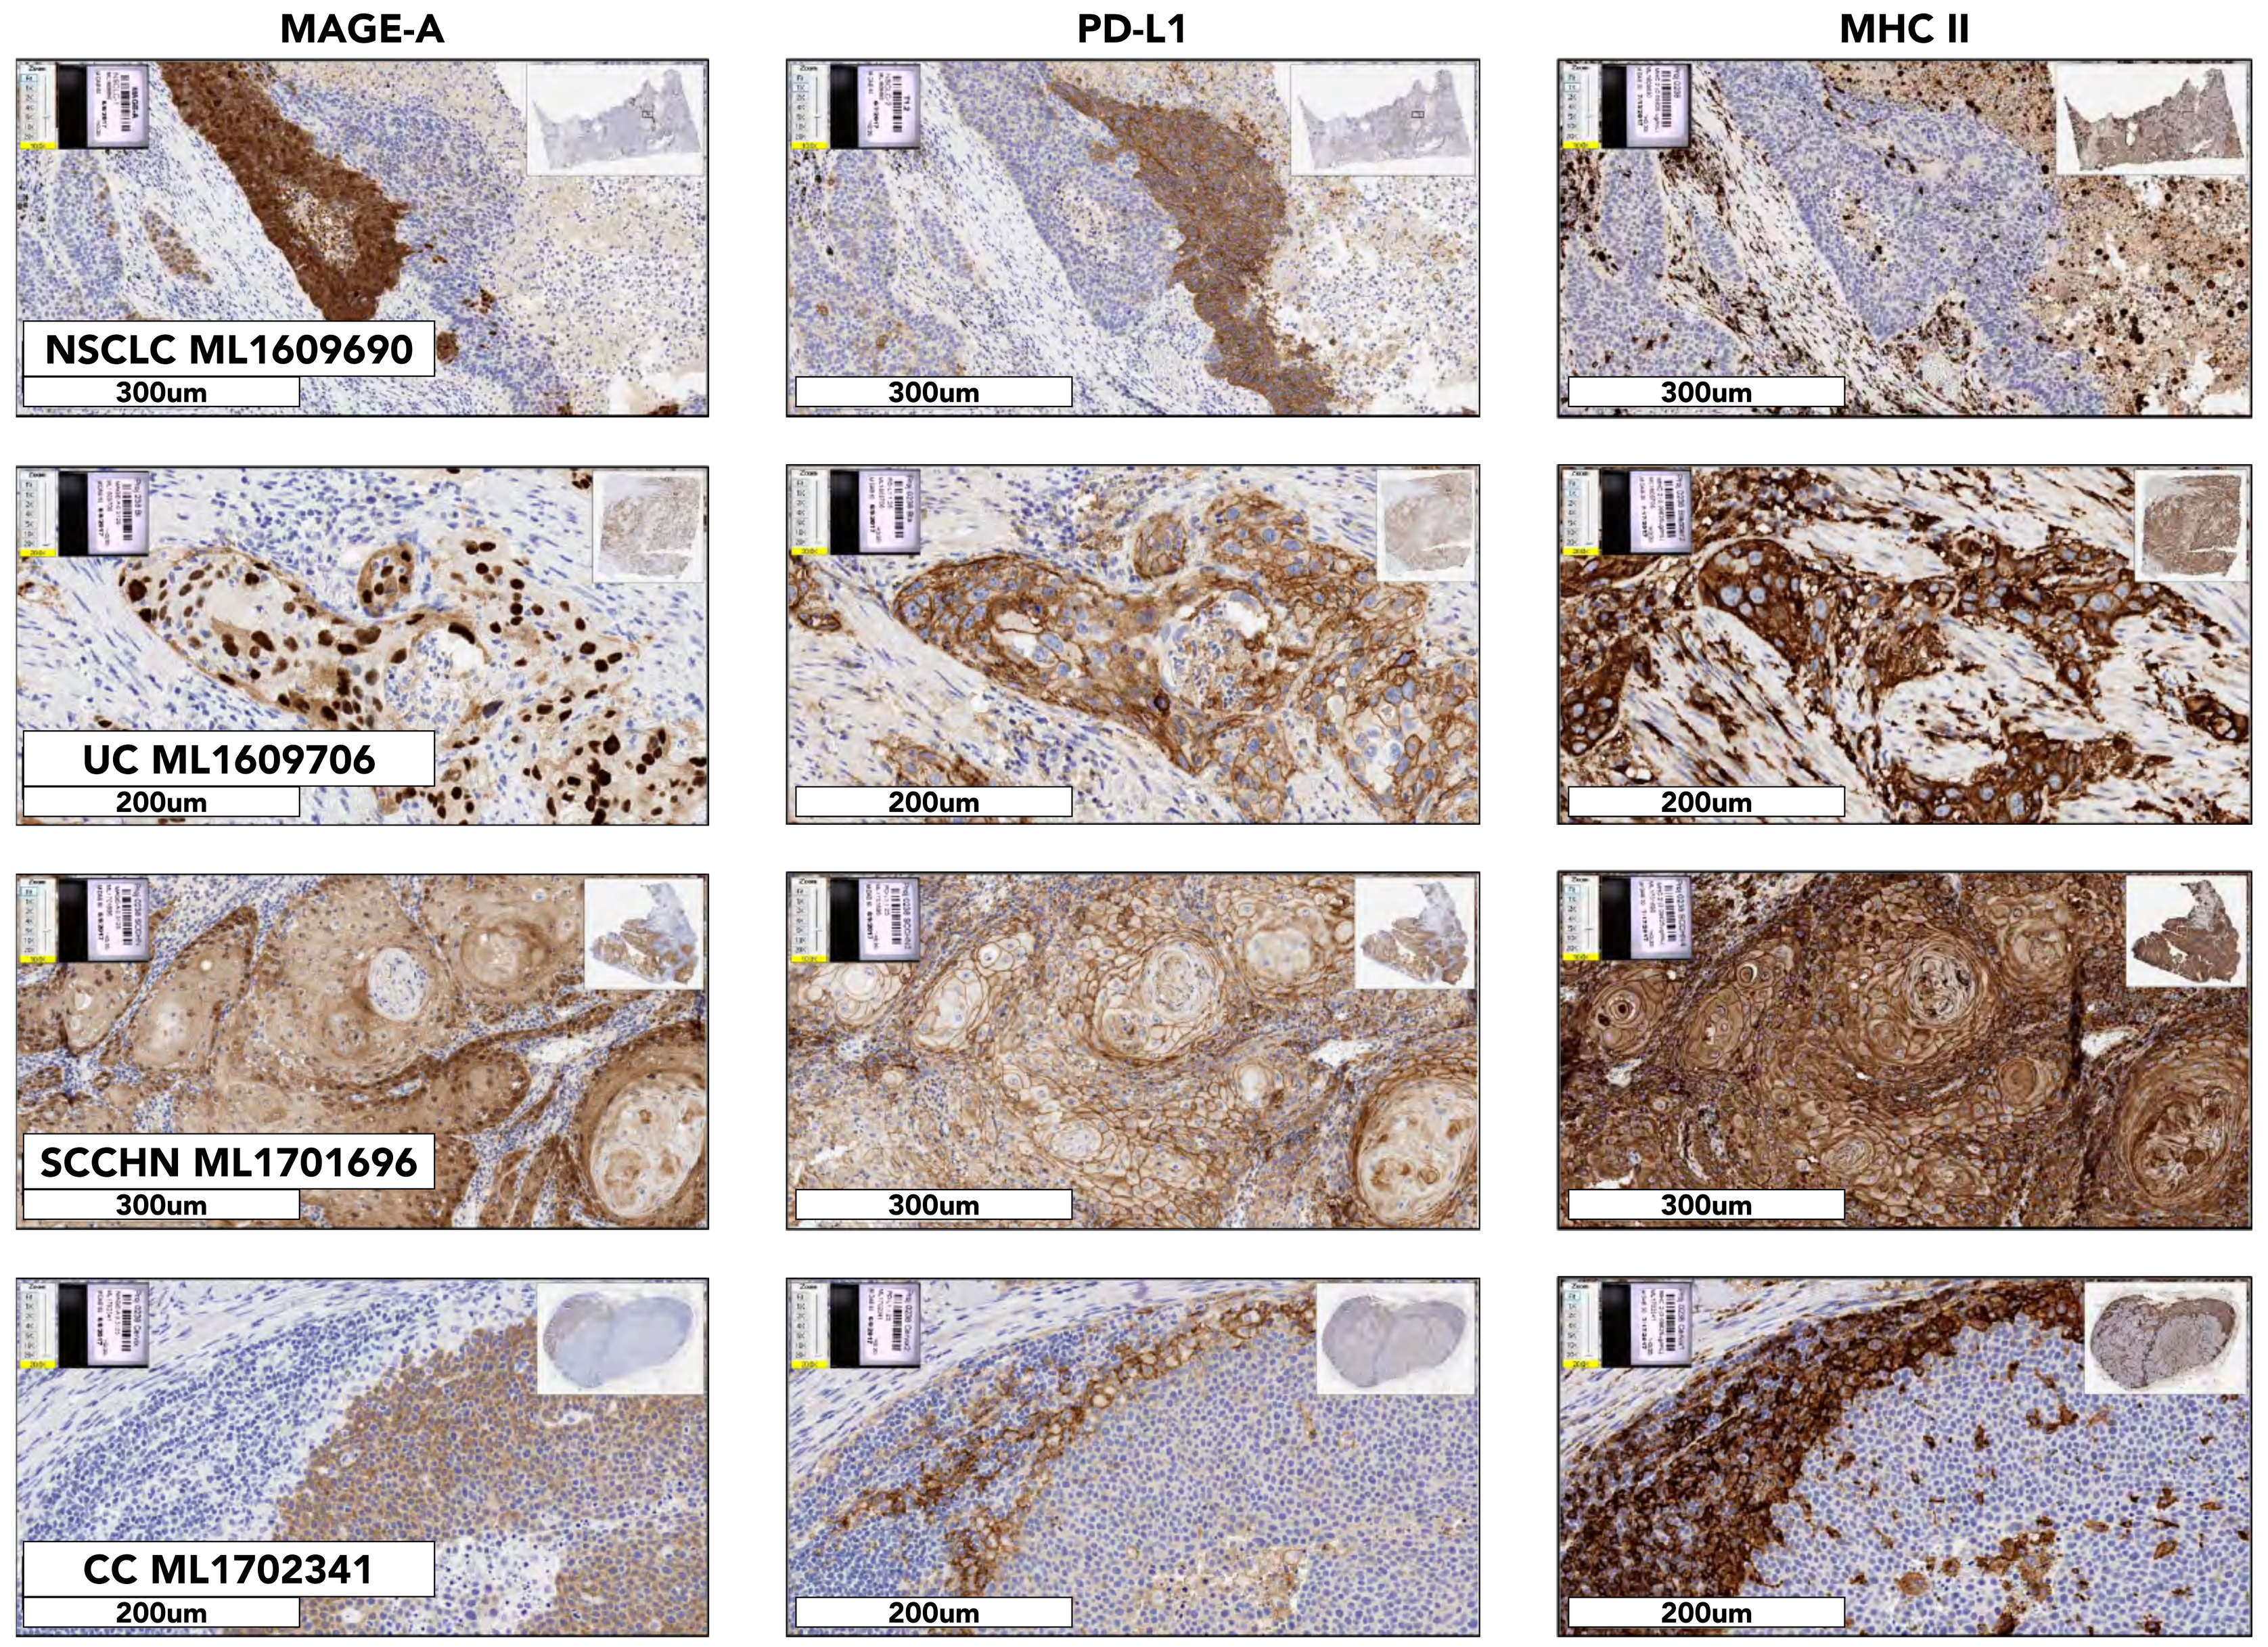

MAGE-A
PD-L1
MHC II
NSCLC ML1609690
UC ML1609706
SCCHN ML1701696
CC ML1702341
300um
200um

Examples of different staining patterns observed in adjacent sections stained for IHC with antibodies to MAGE-A, PD-L1, and MHCII.
CC, cervical cancer; IHC, immunohistochemistry; MAGE, melanoma-associated antigen; MHC, major histocompatibility complex; NSCLC, non-small cell lung cancer; PD-L1, programmed death ligand 1; SCCHN, squamous cell carcinoma of the head and neck; UC, urothelial carcinoma.

Figure 4. MAGE-A3/A6–positive and –negative tumors are similar with respect to PD-L1 gene expression and diversity of immune cell phenotypes including CD4, CD8, Th1, and Treg cells.

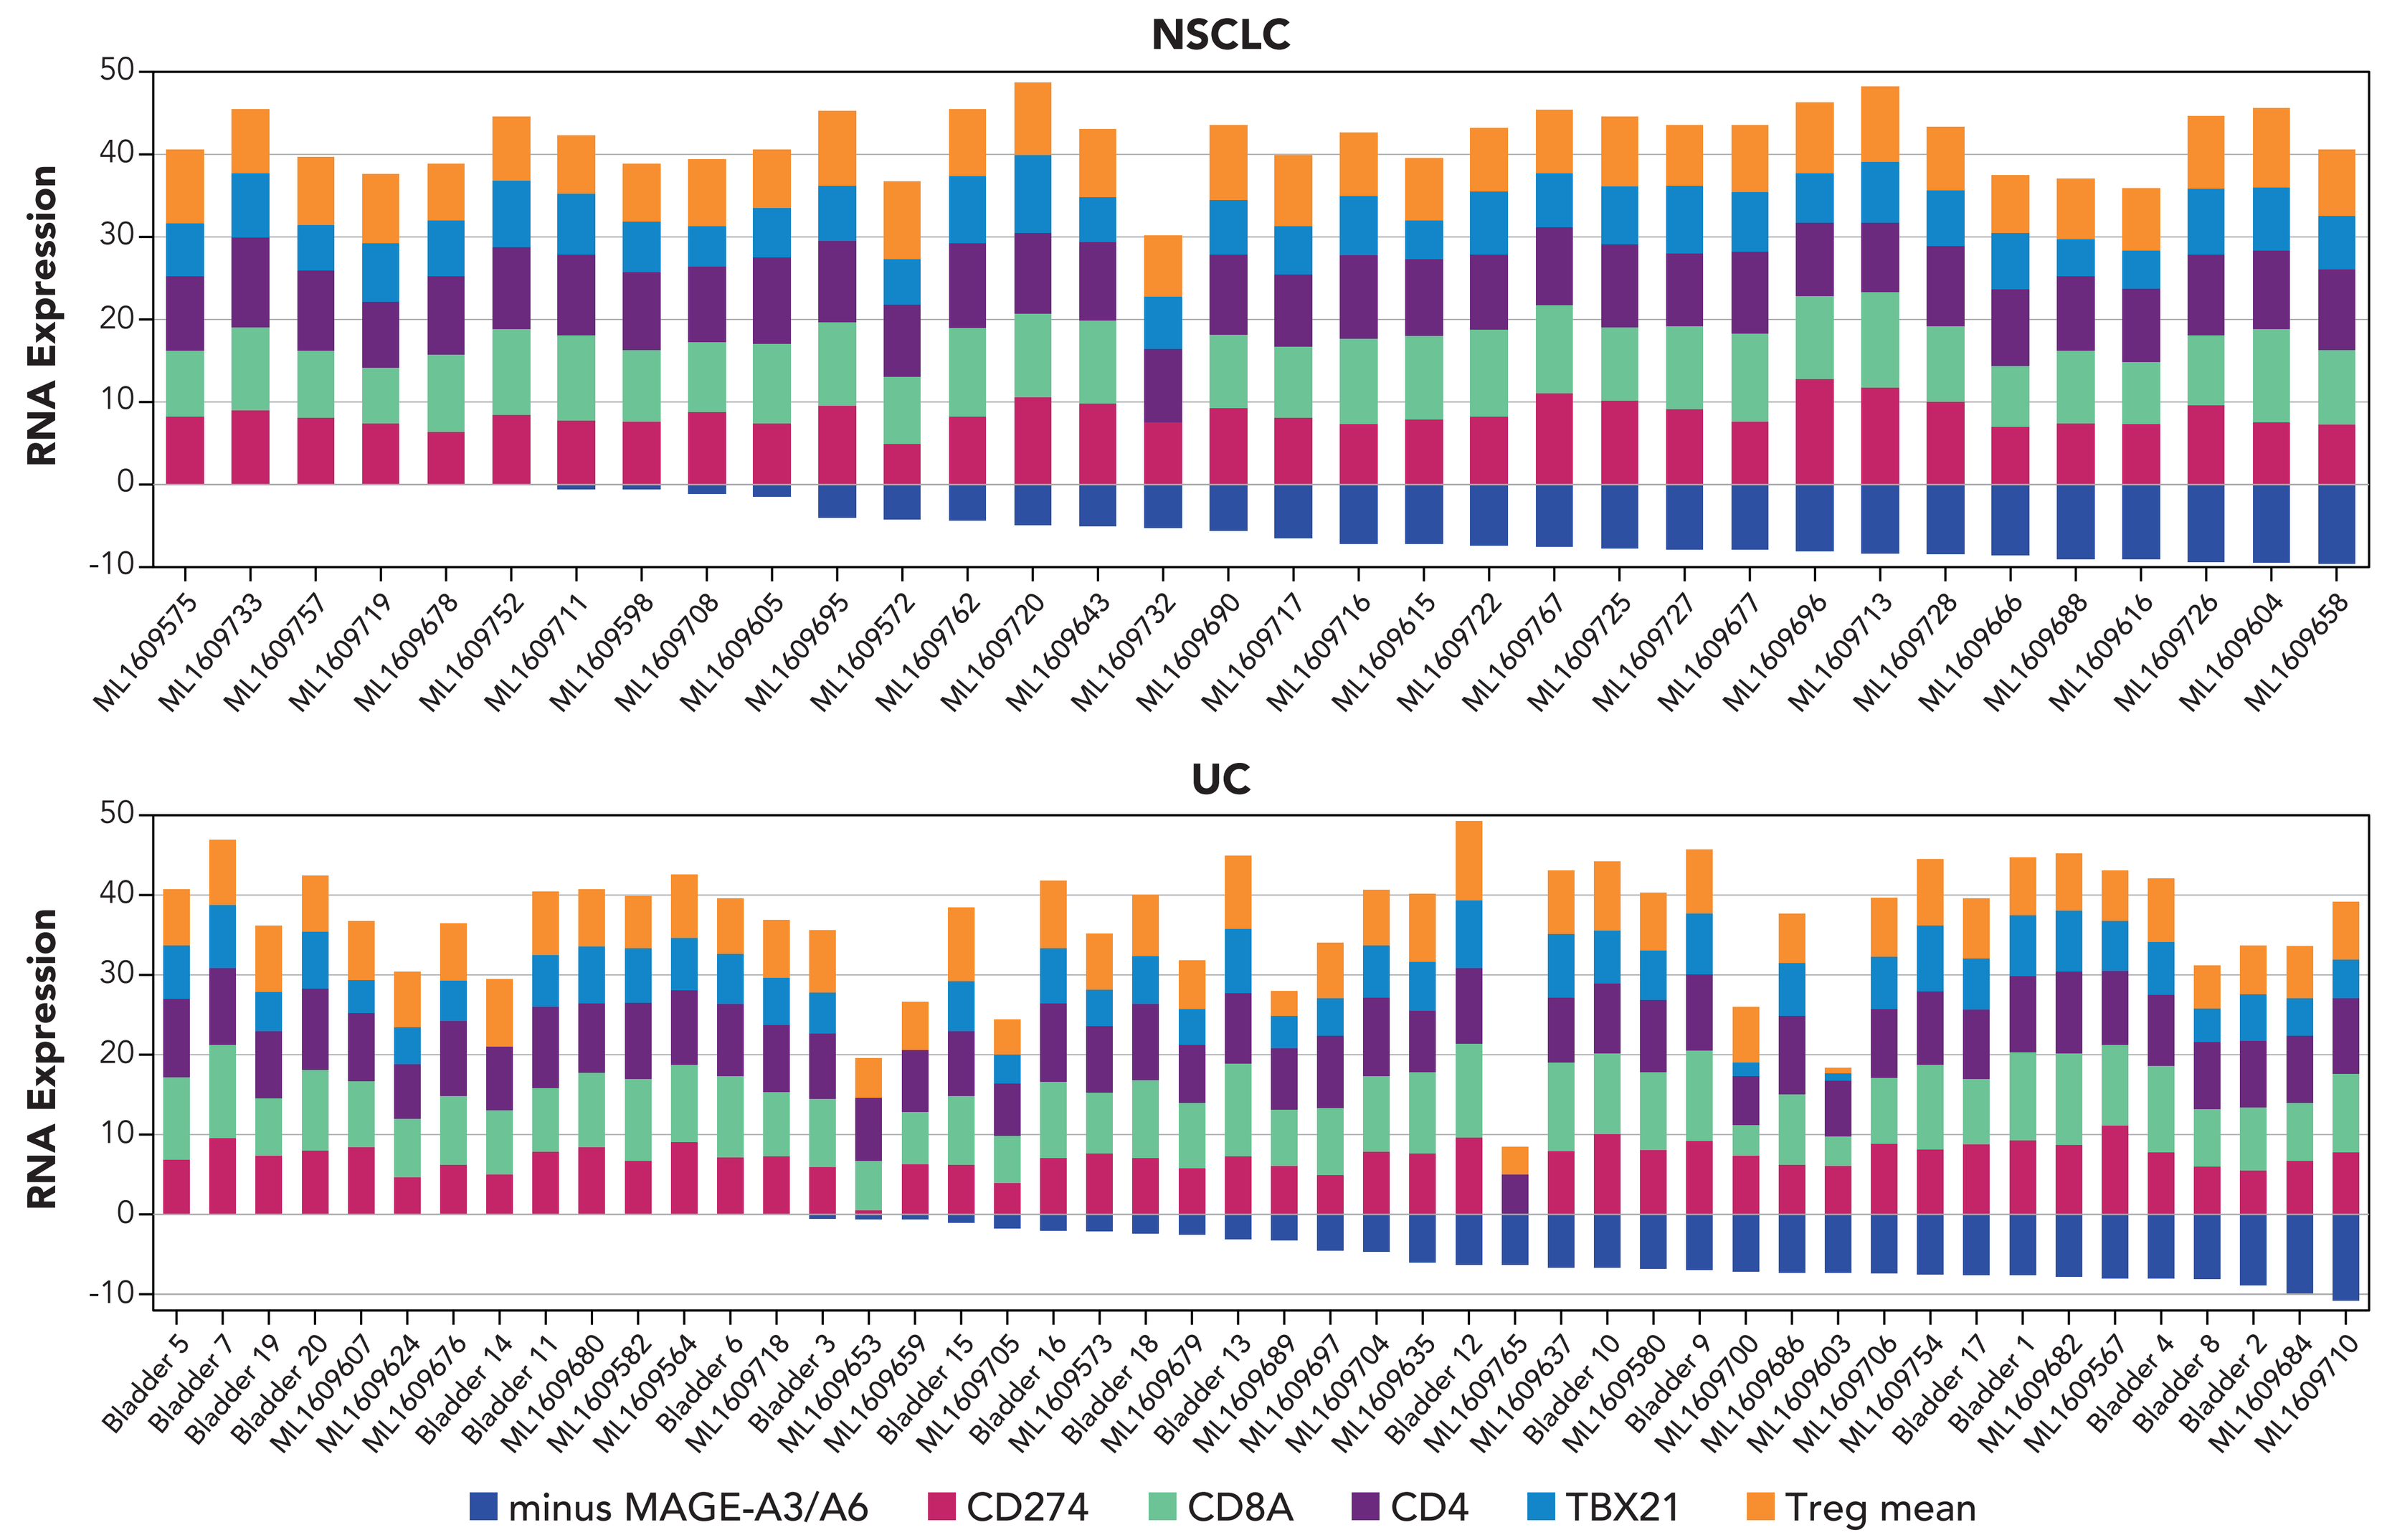

NSCLC
RNA Expression
UC
RNA Expression
minus MAGE-A3/A6
CD274
CD8A
CD4
TBX21
Treg mean

Using values derived from OIRRA, MAGE-A3/6, CD274 (PD-L1), CD4, CD8, TBX21 (marker for Th1), and Treg means (mean of FOXP3 and TNFRSF18) were normalized to RPM and further transformed to LOG2 (RPM+1) format. Forty-eight UC or 34 NSCLC samples were sorted using MAGE-A3/A6 expression from low to high and plotted with expression of CD274, CD4, CD8, TBX21, and regulatory T cells against MAGE-A3/A6 expression (displayed for viewing ease as negative value).
MAGE, melanoma-associated antigen; NSCLC, non-small cell lung cancer; OIRRA, Oncomine™ Immune Response Research Assay; PD-L1, programmed death ligand 1; RPM, reads per million per mapped reads; Th1, type 1 T helper; Treg, regulatory T; UC, urothelial carcinoma.

CONCLUSIONS

- The results support clinical evaluation of a MAGE-A3/A6 TCR T-cell product across these major histologies
- This 2-part MAGE-A3/A6 screening assay may reduce the potential for false-positive results obtained using the 6C1 IHC assay alone and be useful in screening patients for anti–MAGE-A3/A6 T-cell therapy
- Although the significance of the protein and gene expression patterns remain to be determined, the presence of PD-L1–positive tumor cells and immune cells in MAGE-A3/A6–positive tumors supports the concept of checkpoint blockade in combination with T-cell therapy to treat advanced solid tumors

REFERENCES

1. Lu Y-C, et al. *J Clin Oncol*. 2017. epub ahead of print.
2. Kerkar SP, et al. *J Immunother.* 2016;39(4):181-187.
3. Glimcher LH, Kara CJ. *Annu Rev Immunol.* 1992;10:13-49.

ACKNOWLEDGMENTS

Editorial support was provided by Nexus Global Group Science, LLC, funded by Kite, a Gilead company.

DISCLOSURES

SHA, AB: Employment with Kite, a Gilead Company; equity ownership in Gilead. **RJ:** Employment and patent with Kite, a Gilead Company; equity ownership in Gilead, Amgen, Juno, SGEN; **WSC:** Employment with Molecular MD; consultancy with Kite, a Gilead Company; equity in Amgen and Exelixis; **CCF:** Employment with Molecular MD; consultancy with Kite, a Gilead Company; **JL:** Employment with Molecular MD; consultancy with Kite, a Gilead Company; equity ownership in Sanofi; **IF and AD:** None.